POSTER FRI-376

An Open-Label Study to Assess the Safety and Efficacy of COR-003 (Levoketoconazole) in the Treatment of Endogenous Cushing's Syndrome

Roberto Salvatori, MD[1], Anthony DelConte, MD[2], Eliza B Geer, MD[3], Theodore R Koziol, PhD[4], Diane Jorkasky, MD[5]

BACKGROUND

- Endogenous Cushing's syndrome (CS) is a rare disorder[1]
- Approved medical therapy options for the management of hypercortisolism in CS are limited, and no US Food and Drug Administration–approved treatment reduces cortisol levels by acting directly on the adrenal glands[2-5]
- Ketoconazole, a racemic mixture of 2 enantiomers (2R,4S and 2S,4R) and an inhibitor of cortisol synthesis, has been used off-label for many years as an effective treatment for CS (Figure 1)[3,6,7]

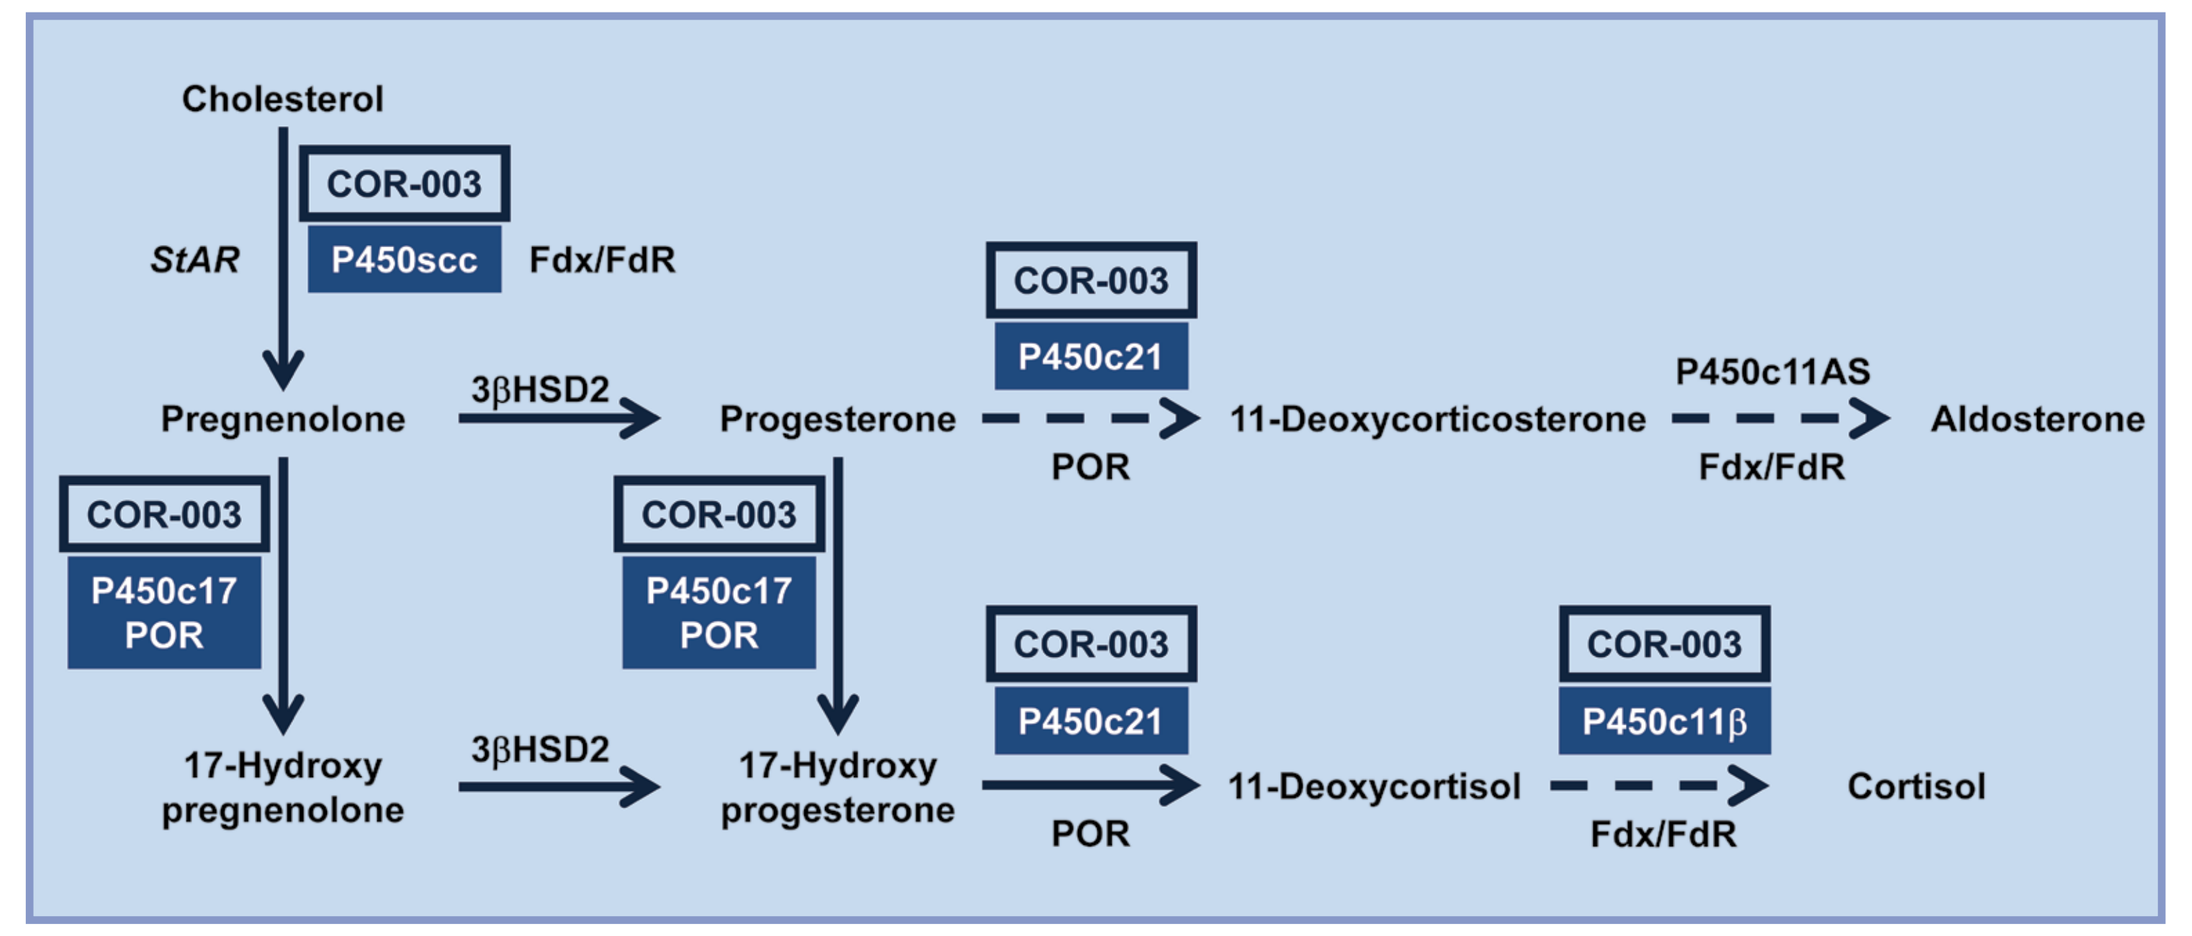
Cholesterol
COR-003
StAR
P450scc
Fdx/FdR
Pregnenolone
3βHSD2
Progesterone
COR-003
P450c21
POR
11-Deoxycorticosterone
P450c11AS
Fdx/FdR
Aldosterone
COR-003
P450c17
POR
COR-003
P450c17
POR
17-Hydroxy pregnenolone
3βHSD2
17-Hydroxy progesterone
COR-003
P450c21
POR
11-Deoxycortisol
COR-003
P450c11β
Fdx/FdR
Cortisol

3βHSD, 3-beta-hydroxysteroid dehydrogenase; FdR, ferredoxin reductase; Fdx, ferredoxin; POR, P450 oxidoreductase; StAR, steroidogenic acute regulatory protein.

[8]Adapted from Miller WL, Auchus R. *Endocr Rev.* 2011;32(1):81-151.

Figure 1. Pathway of Corticosteroid Synthesis and Inhibition by COR-003 (Levoketoconacole)

COR-003 (Levoketoconazole)

- Cortendo AB is developing COR-003, the single 2S,4R enantiomer of racemic ketoconazole, as an investigational new drug for the treatment of endogenous CS
- COR-003 is developed as an oral, twice-daily formulation
- The SONICS study (ClinicalTrials.gov, NCT01838551) is a pivotal phase 3 study to investigate the safety and efficacy of COR-003 in the treatment of endogenous CS

STUDY DESIGN

- Single-arm, open-label study to assess the efficacy, safety, tolerability, and pharmacokinetics (PK) of COR-003
- Following screening, there are 3 treatment phases: dose titration, maintenance (6 months at therapeutic dose), and extended evaluation (6 months of continued treatment after maintenance) (Figure 2)

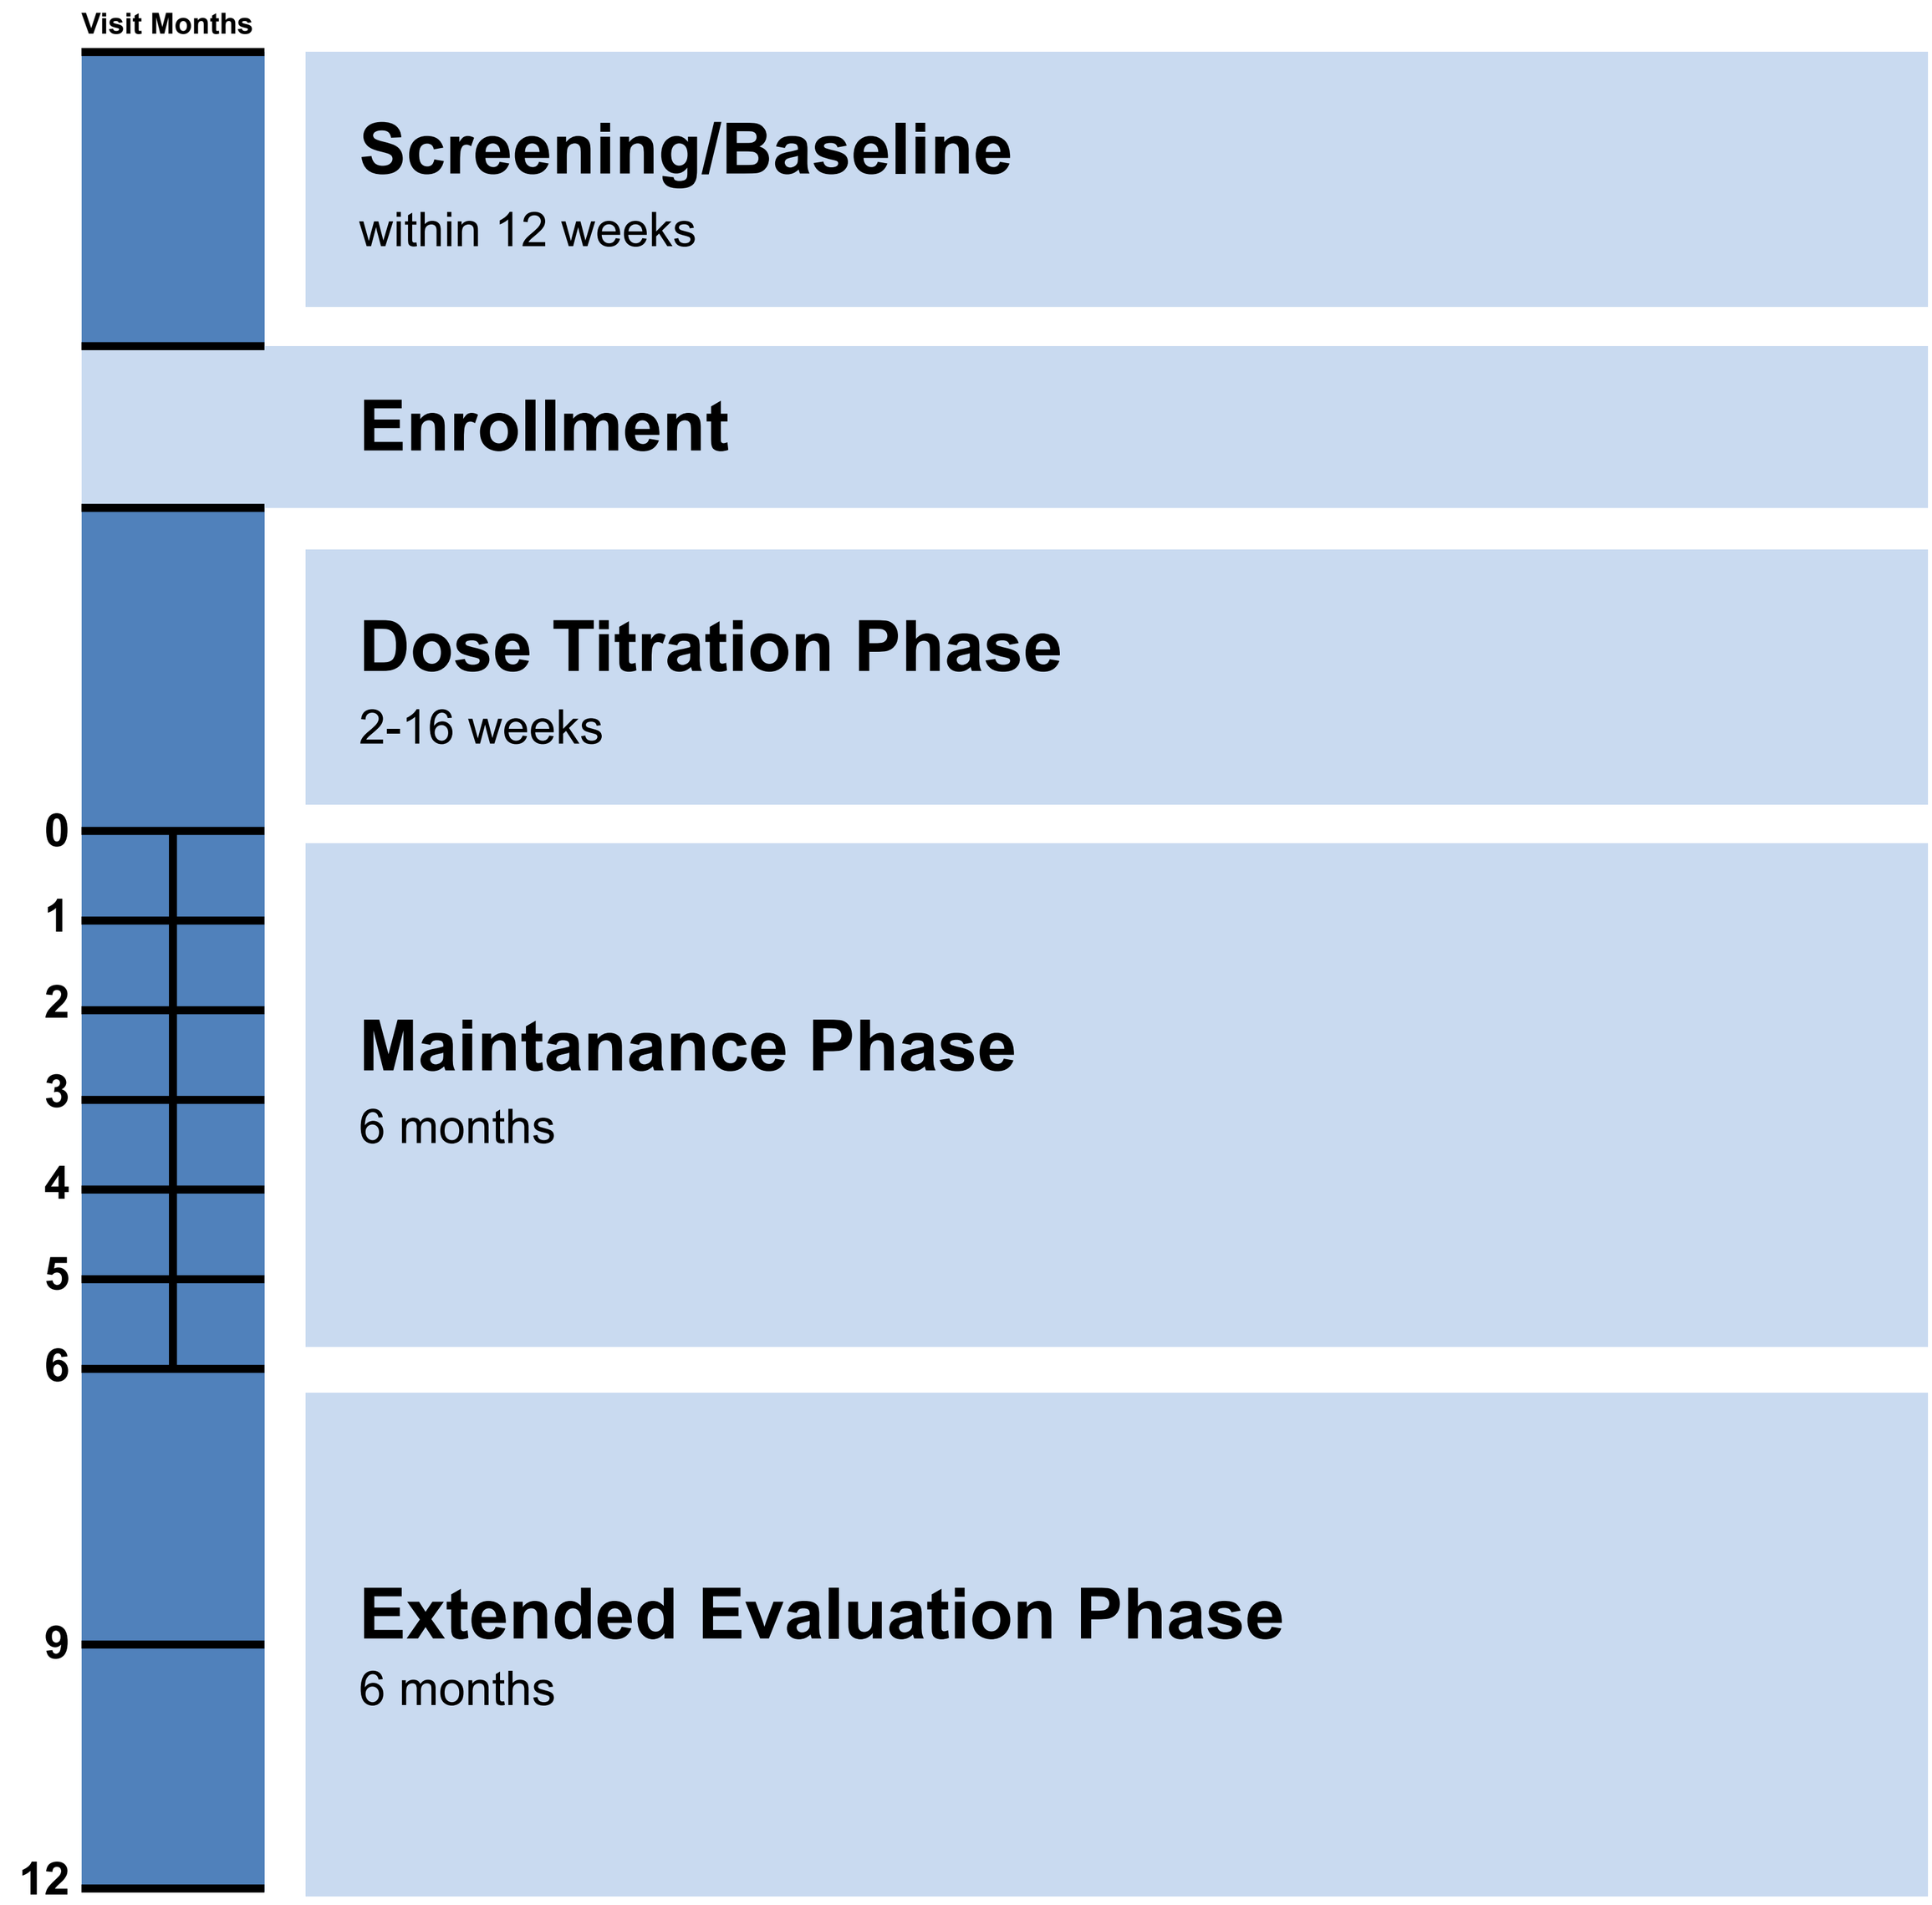
Visit Months
Screening/Baseline
within 12 weeks
Enrollment
Dose Titration Phase
2-16 weeks
0
1
2
3
4
5
6
9
12
Maintanance Phase
6 months
Extended Evaluation Phase
6 months

Figure 2. Study Design

- During the dose-titration phase, COR-003 is administered twice daily, and the dose is increased until:
 - Mean 24-hour urinary free cortisol (UFC) levels are less than or equal to the upper limit of normal (≤ULN) as assessed by a central laboratory, or
 - The highest tolerated dose has been reached
- Patients then enter the maintenance phase and return monthly for 6 months for evaluation of efficacy (by UFC measurements), and for clinical, safety, metabolic, quality of life, and PK assessments
- During extended evaluation, patients will return every 3 months for assessments (ie, during months 9 and 12) and will undergo interim laboratory evaluations

STUDY METHODOLOGY

Study Eligibility

- Selected inclusion criteria:
 - Patients aged ≥18 years with a confirmed diagnosis of persistent or recurrent CS (with or without previous therapy) or newly diagnosed disease (excluding adrenal carcinoma) who are not candidates for surgery
- Selected exclusion criteria:
 - Nonendogenous source of hypercortisolemia (eg, exposure to exogenous glucocorticoids)
 - Pseudo-Cushing's syndrome (based on investigator assessment)
 - De novo Cushing's disease and a candidate for surgery
 - Treated with radiation within the previous 4 years or >4 years with evidence of improvement in disease within 6 months prior to screening

Primary Endpoint

- Reduction in mean 24-hour UFC levels to ≤ULN following 6 months of maintenance phase therapy without additional dose increase

Selected Secondary Endpoints

- Monthly assessments throughout 6 months of dosing in the maintenance phase and at 9 and 12 months in the extended evaluation phase to include mean UFC levels and mean plasma and late-night salivary cortisol levels
- Clinical signs and symptoms, HbA1c and glucose levels, blood pressure values, lipid profile results, C-reactive protein level, weight, and quality of life
- Safety evaluations (clinical observations and adverse event reporting)

Study Assessments

- Demographic/medical history
- Efficacy (24-hour urine and midnight salivary cortisol collections)
- Assessment of clinical signs and symptoms of CS
- Quality of life assessments
- Safety (eg, physical examination, vital signs, plasma adrenocorticotropic hormone levels, pituitary magnetic resonance imaging)
- PK sample collection

Data Analysis

- Analyses will be conducted on the intent-to-treat (ITT) and per protocol (PP) populations
 - ITT population (primary analysis population): all patients who receive ≥1 dose of COR-003, who will be used to evaluate efficacy and safety
 - PP population: all patients who enter the maintenance phase of the study, receive ≥80% of planned doses, and have no major protocol deviations

Statistical Considerations

- Proportion of responders in the ITT and PP populations will be estimated along with corresponding 95% confidence intervals (CIs); if the lower bound of the 95% CI in the ITT population is ≥20%, COR-003 will be considered to be effective

Safety Summary Analyses

- Study drug exposure: average daily and cumulative dose, and total days on study drug
- Adverse events: all, most common, drug-related, serious, and those leading to discontinuation and withdrawal from study

DISCUSSION

- There is a need for new medical therapy options to manage hypercortisolism in CS[2-5]
- Ketoconazole has a long history of effective off-label use in the treatment of CS but is associated with potential hepatotoxicity[3,6,7]
- The SONICS trial investigating the efficacy and safety of COR-003, the 2S,4R enantiomer of ketoconazole, is currently enrolling patients with endogenous CS
- In vitro, COR-003 has a higher potency for the inhibition of cortisol biosynthesis enzymes and a considerably lower potency to inhibit Cyp7A1,[9] a key enzyme for bile acid synthesis, compared with ketoconazole and may therefore have a higher therapeutic index
- As observed with the off-label use of ketoconazole in CS,[6] COR-003, the single enantiomer of ketoconazole, may have beneficial effects in secondary endpoints contributing to morbidity and mortality in patients with CS, including hyperglycemia, dyslipidemia, obesity, and hypertension

Study funded by Cortendo AB. Dr Salvatori and Dr Geer are investigators in the trial sponsored by Cortendo AB. Dr DelConte, Dr Koziol, and Dr Jorkasky are paid consultants to Cortendo AB. Connexion Healthcare provided writing and editorial assistance that was supported by funding from Cortendo AB.

References

1. Badia X, Valassi E, Roset M, Webb S. Disease-specific quality of life evaluation and its determinants in Cushing's syndrome: what have we learnt? *Pituitary.* 2014;17:187-195.
2. Feelders RA, Hofland LJ. Medical treatment of Cushing's disease. *J Clin Endocrinol Metab.* 2013;98(2):425-438.
3. Cuevas-Ramos D, Fleseriu M. Treatment of Cushing's disease: a mechanistic update. *J Endocrinol.* 2014;223(2):R19-R39.
4. Signifor (pasireotide) [package insert]. East Hanover, NJ: Novartis Pharmaceuticals Corporation; 2012.
5. Korlym (mifepristone) [package insert]. Menlo Park, CA: Corcept Therapeutics; 2013.
6. Castinetti F, Guignat L, Giraud P. Ketoconazole in Cushing's disease: is it worth a try? *J Clin Endocrinol Metab.* 2014;99:1623-1630.
7. Engelhardt D, Mann K, Hörmann R, et al. Ketoconazole inhibits cortisol secretion of an adrenal adenoma in vivo and in vitro. *Klin Wochenschr.* 1983;61(7):373-375.
8. Miller WL, Auchus RJ. The molecular biology, biochemistry, and physiology of human steroidogenesis and its disorders. *Endocr Rev.* 2011;32(1):81-151.
9. Rotstein DM, Kertesz DJ, Walker KAM, Swinney DC. Stereoisomers of ketoconazole: preparation and biological activity. *J Med Chem.* 1992;35(15):2818-2825.

[1]Johns Hopkins University, Baltimore, MD. [2]Saint Joseph's University, Philadelphia, PA. [3]Mount Sinai Medical Center, New York, NY. [4]Consultant to Cortendo AB, Radnor, PA. [5]University of California San Francisco, San Francisco, CA.

ENDO 2015 | March 5-8, 2015 | San Diego, CA