Long-Term Efficacy and Adverse Event Characterization of Dichlorphenamide for the Treatment of Primary Periodic Paralysis

Nicholas E. Johnson, MD[1]; Michael G. Hanna, MD[2]; Jeffrey M. Statland, MD[3]; Fredric Cohen, MD[4]; Robert C. Griggs, MD[5]

[1]Virginia Commonwealth University, Richmond, VA, USA; [2]MRC Centre for Neuromuscular Diseases, UCL Institute of Neurology, London, UK; [3]University of Kansas Medical Center, Kansas City, KS, USA; [4]Strongbridge Biopharma, Trevose, PA, USA; [5]University of Rochester Medical Center, Rochester, NY, USA

Poster #102

Introduction

- Primary periodic paralysis (PPP) is a rare condition caused by genetic mutations in skeletal muscle sodium, calcium, and potassium channels, resulting in attacks or muscle weakness[1,2]
- Dichlorphenamide (DCP), administered daily for up to 1 year, has been shown to reduce the frequency, severity, and duration of attacks of PPP[3,4] and was approved in the United States for the treatment of primary hyperkalemic and hypokalemic periodic paralysis and related variants in 2015[5]
- The most common adverse events (AEs) observed during the hyperkalemic PPP (HYP)/hypokalemic PPP (HOP) trial were paresthesia and cognition-related AEs[3,4]
 - In the double-blind phase of the HYP/HOP trial (n=65), compared with placebo, DCP-treated patients had higher rates of paresthesia (placebo, 14% vs DCP, 47%) and cognitive disorder (placebo, 7% vs DCP, 19%)[3]

Objective

- To evaluate long-term (9 weeks blinded plus 52 weeks open-label) efficacy and characterize paresthesia and cognition-related AEs in a pooled HYP/HOP analysis

Methods

Study Design and Patients

- Data were analyzed from a 9-week phase 3, randomized, double-blind, placebo (PBO)-controlled phase and a 52-week open-label extension phase (**Figure 1**)[3]
 - (Planned) acetazolamide (ACZ) arm was dropped due to low enrollment (n=5)
- Adults in the HYP/HOP trial were randomly assigned to receive DCP (current DCP dose if taking DCP before study start or DCP 50 mg twice daily; patients receiving ACZ before study start were assigned a DCP dose equivalent to 20% of the ACZ dose) or PBO for 9 weeks during the double-blind phase, followed by a 52-week open-label phase (**Figure 1**)[3]
 - Patients in the open-label DCP group continued at the same dose they were taking at the end of the 9-week double-blind phase (with dose adjustments permitted to mimic clinical practice; DCP/DCP population)
 - Patients in the PBO group switched to DCP 50 mg twice daily in the open-label phase (PBO/DCP population)

Figure 1. Study Design*

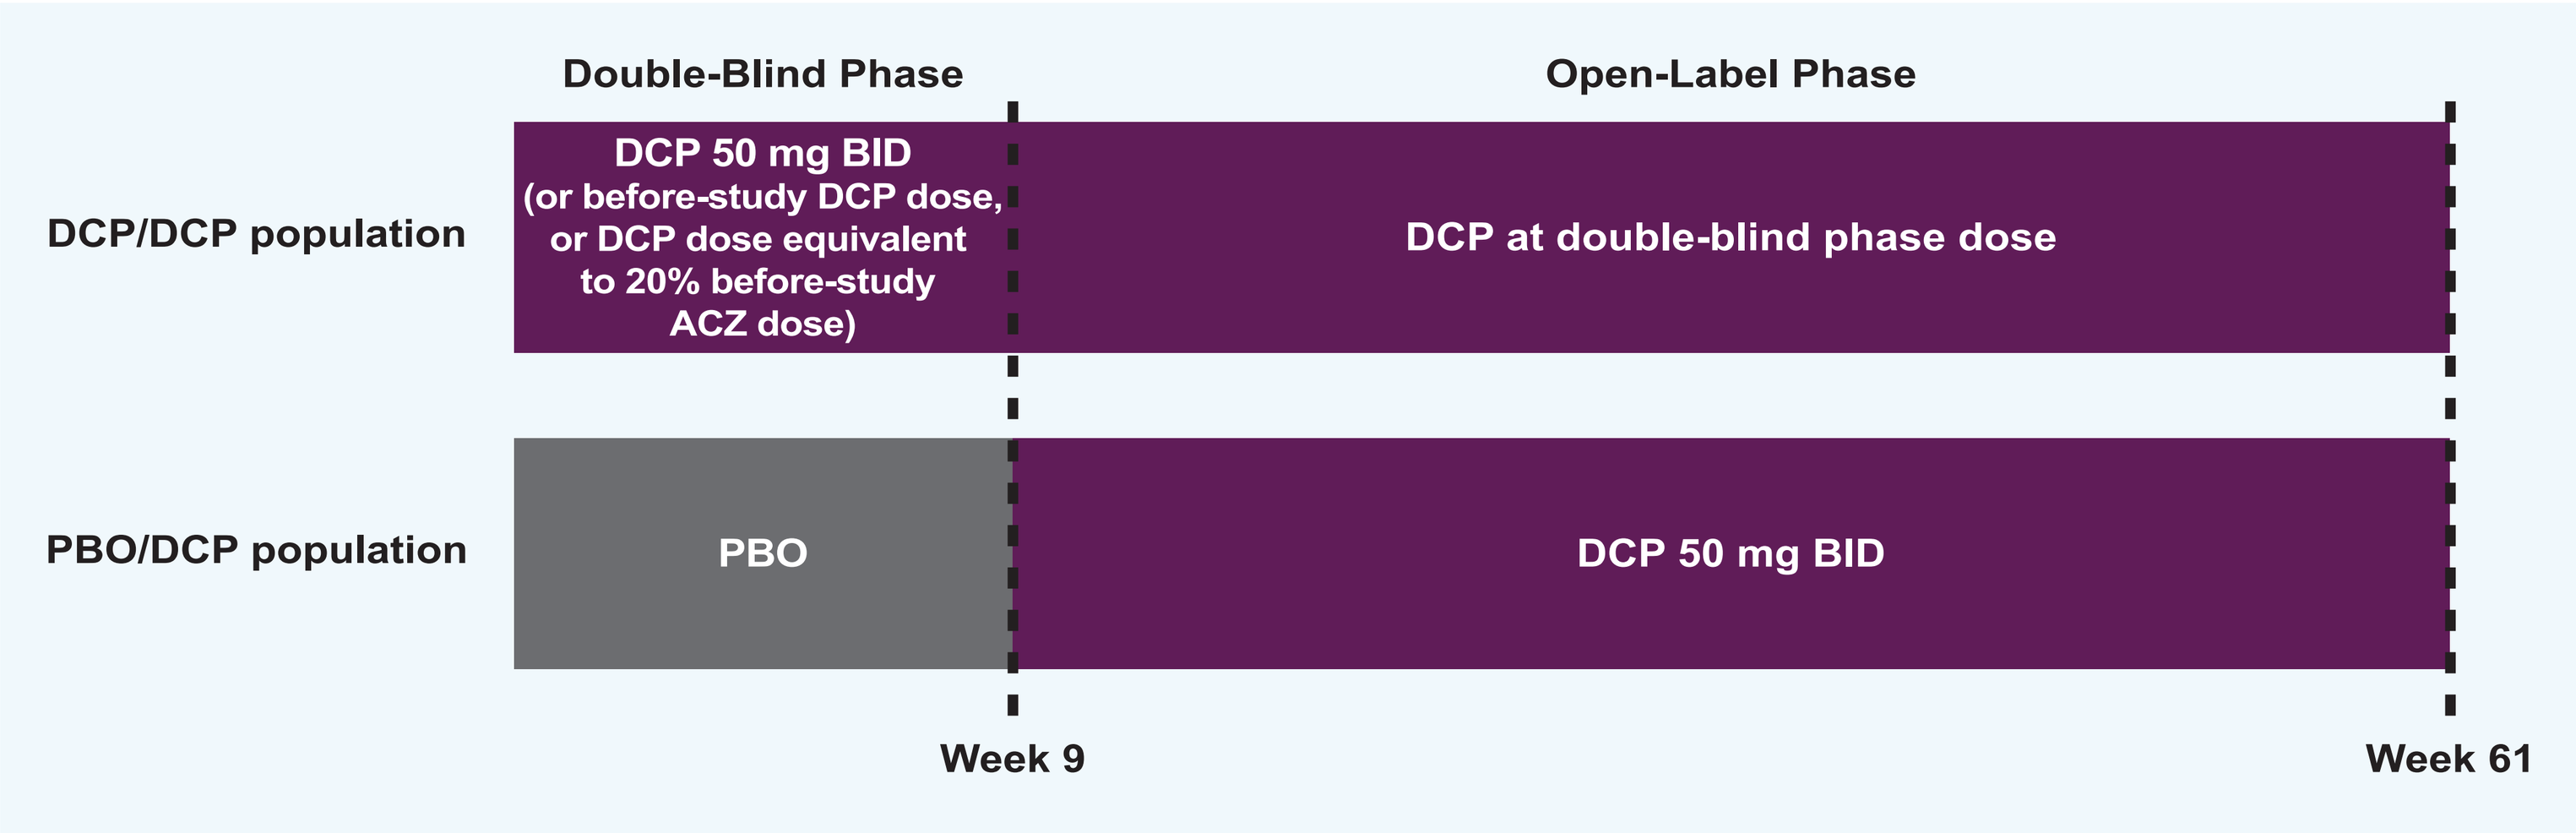

Double-Blind Phase
Open-Label Phase
DCP/DCP population
DCP 50 mg BID (or before-study DCP dose, or DCP dose equivalent to 20% before-study ACZ dose)
DCP at double-blind phase dose
PBO/DCP population
PBO
DCP 50 mg BID
Week 9
Week 61

*Information from Sansone VA, et al. *Neurology*. 2016;86(15 Suppl):1408-1416.[3]
ACZ = acetazolamide; BID = twice daily; DCP = dichlorphenamide; PBO = placebo.

Assessments and Statistical Analyses

- The study completer population included patients in the intent-to-treat (ITT) population (all patients randomly assigned in the double-blind phase to DCP or PBO) who completed the open-label phase of the study
- Changes from baseline to Week 61 in attack rates and severity-weighted attack rates were analyzed for within- and between-group median differences
 - Within-group median differences were assessed for significance using Wilcoxon signed rank test; between-treatment differences were assessed using blocked Wilcoxon rank sum test, adjusting for type of PPP
- AEs reported during DCP exposure were summarized by double-blind/open-label treatment (PBO/DCP or DCP/DCP) groups
 - Medical Dictionary for Regulatory Activities high level group terms of deliria (including confusion) and mental impairment disorders were pooled for cognition-related events

Results

Demographic and Baseline Characteristics

- 63 patients were included in the ITT population (DCP/DCP, n=36; PBO/DCP, n=27; **Table 1**)
 - 2 of 65 patients in the PBO group discontinued from study before the open-label extension phase
 - Majority of the 63 patients were male (61.9%), white (84.1%), and had hypokalemic periodic paralysis (68.3%)
- 47 patients (74.6%) completed 61 weeks of treatment (study completer population)

Table 1. Demographic and Baseline Disease Characteristics

Parameter	ITT Population		Study Completer Population	
	DCP/DCP (N=36)	PBO/DCP (N=27)	DCP/DCP (N=26)	PBO/DCP (N=21)
Age, y				
Mean (SD)	42.9 (13.3)	45.2 (15.6)	40.9 (14.2)	43.4 (15.9)
Range	19–76	19–76	19–76	19–76
Male, n (%)	22 (61.1)	17 (63.0)	17 (65.4)	13 (61.9)
Race, n (%)				
White	30 (83.3)	23 (85.2)	22 (84.6)	17 (81.0)
Hispanic	4 (11.1)	2 (7.4)	3 (11.5)	2 (9.5)
Other	2 (5.6)	1 (3.7)	1 (3.8)	1 (4.8)
Not reported	0	1 (3.7)	0	1 (4.8)
Type of periodic paralysis, n (%)				
Hyperkalemic	12 (33.3)	8 (29.6)	9 (34.6)	7 (33.3)
Hypokalemic	24 (66.7)	19 (70.4)	17 (65.4)	14 (66.7)
Median weekly attack rate	1.75	2.25	1.75	3.00

DCP = dichlorphenamide; ITT = intent-to-treat; PBO = placebo; SD = standard deviation.

Efficacy

- For patients in the study completer population, the median weekly attack rate (**Table 2**; **Figure 2**) and median severity-weighted attack rate (**Table 2**) improved significantly from baseline to Week 61 in both treatment groups

Table 2. Summary of Efficacy From Baseline to Week 61

Outcome	ITT Population		Study Completer Population	
	DCP/DCP (N=36)	PBO/DCP (N=27)	DCP/DCP (N=26)	PBO/DCP (N=21)
Median weekly attack rate				
Baseline	1.75*	2.25†	1.75‡	3.00§
Week 61			0.06	0.25
Median decrease from baseline to Week 61			1.00	0.63
Median % decrease from baseline to Week 61			93.8	75.0
P value¶			<0.0001	0.01
Median severity-weighted attack rate				
Baseline	3.25*	5.88†	2.25‡	5.88§
Week 61			0.06	0.50
Median decrease from baseline to Week 61			2.25	1.69
Median % decrease from baseline to Week 61			97.1	80.8
P value¶			<0.0001	0.01

*n=35. †n=26. ‡n=25. §n=20. ¶Based on median decrease from baseline to Week 61 within a group.
DCP = dichlorphenamide; ITT = intent-to-treat; PBO = placebo.

- Between-group differences in median weekly attack rates (**Table 2**; **Figure 2**) and median severity-weighted weekly attack rates (**Table 2**) were not significant for changes from baseline at Week 61

Figure 2. Median Weekly Attack Rates at Baseline, Week 9, and Week 61 (Study Completer Population)

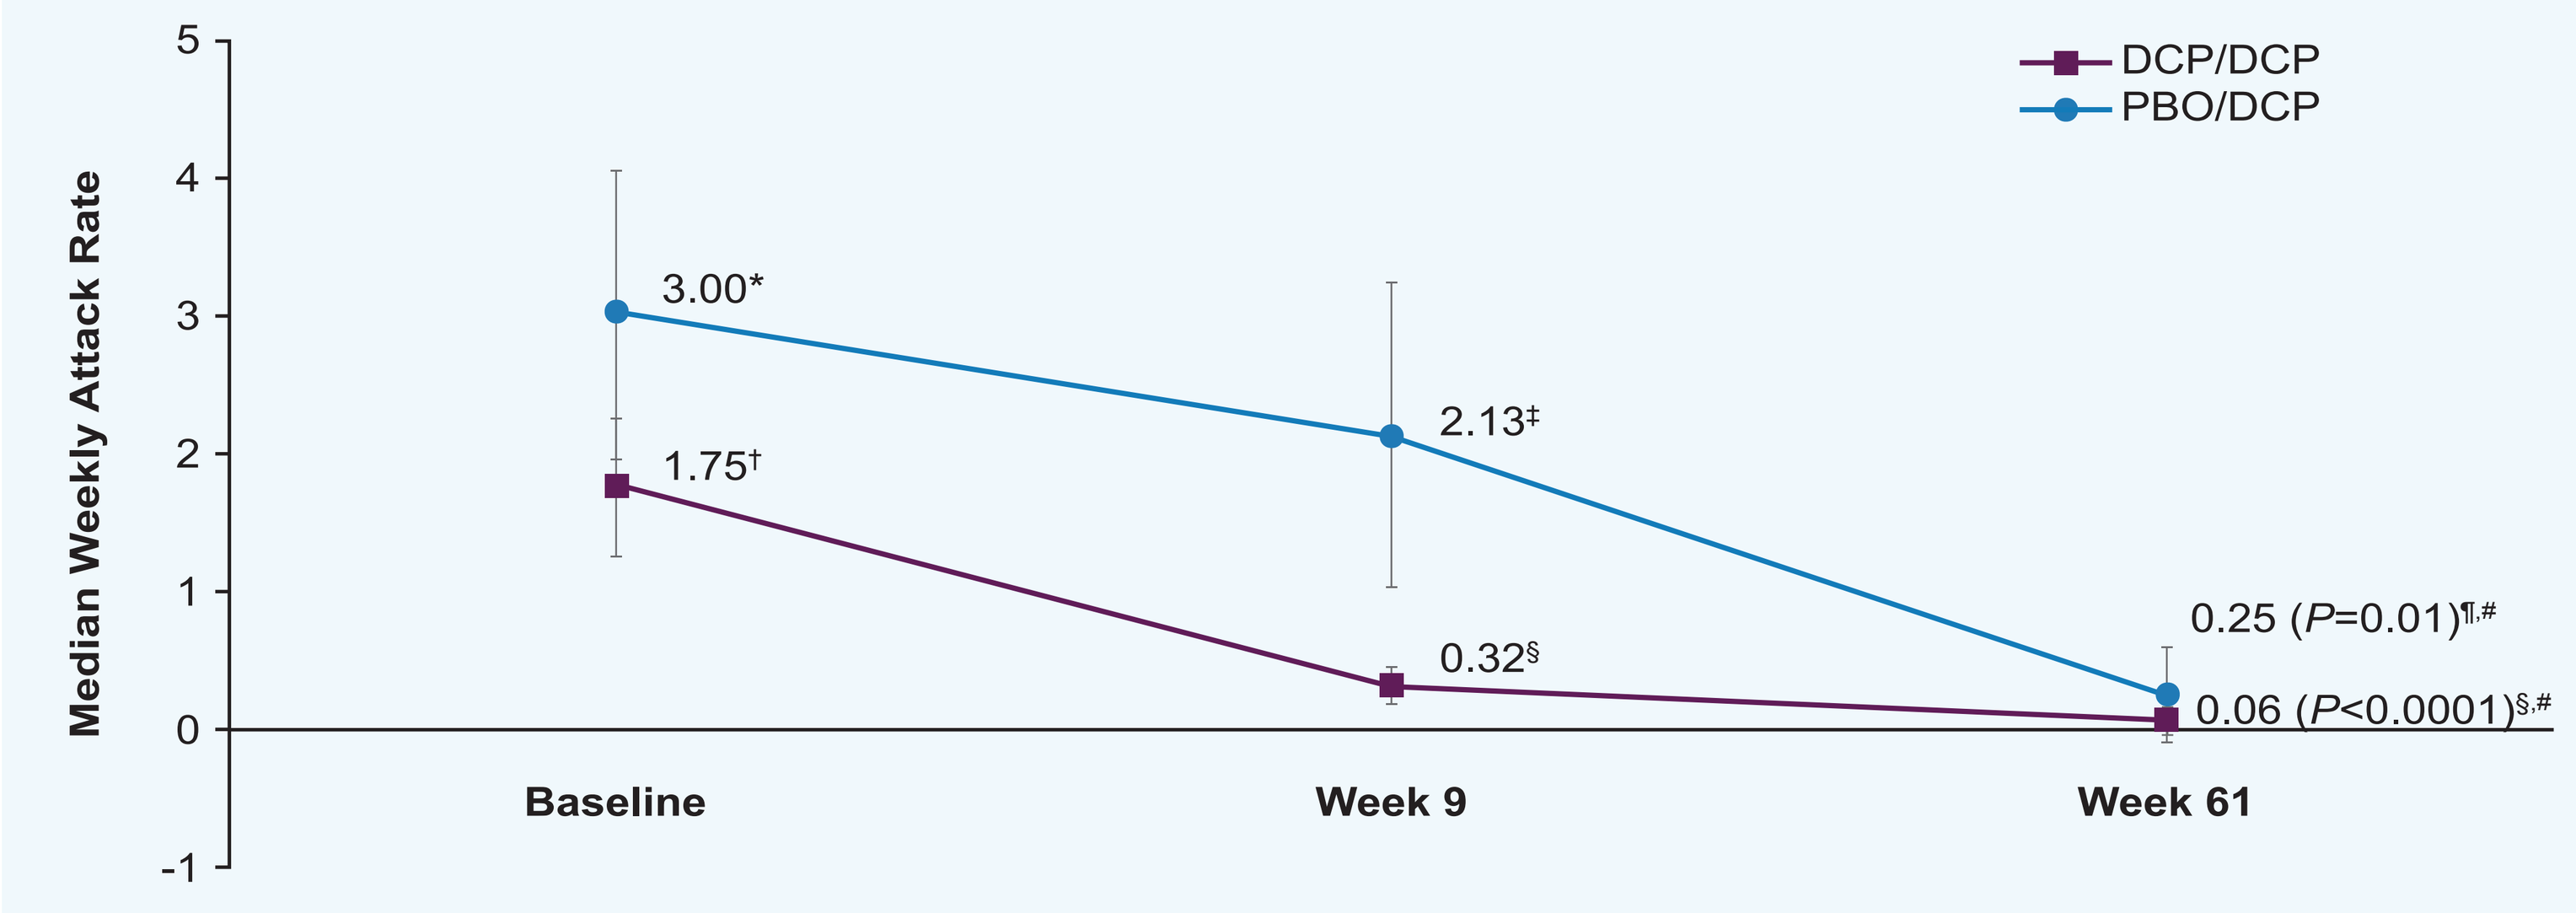

DCP/DCP
PBO/DCP
Median Weekly Attack Rate
5
4
3
2
1
0
-1
3.00*
1.75†
2.13‡
0.32§
0.25 (P=0.01)¶,#
0.06 (P<0.0001)§,#
Baseline
Week 9
Week 61

Bars represent standard error. Attack rates at Week 9 and Week 61 are the mean of the weekly attack rates at Weeks 2–9 and Weeks 54–61, respectively. For patients with <8 weeks of completed diary entries, missing data were considered as days without an attack in a particular week.
*n=20. †n=25. ‡n=16. §n=26. ¶n=21. #*P* value based on median decrease from baseline to Week 61 within a group.
DCP = dichlorphenamide; PBO = placebo.

Paresthesia and Cognition-Related Adverse Events

- Overall, 39.7% (25/63) and 25.4% (16/63) of patients treated with DCP had ≥1 paresthesia and cognition-related AEs, respectively, during the 61-week study duration (**Table 3**)
 - No AEs of paresthesia were considered severe in intensity and only 1 patient discontinued from the study due to paresthesia
 - The majority of cognition-related AEs were mild in intensity and 4 patients (6.3%) discontinued from the study due to a cognition-related AE
- 24.0% (6/25) and 37.5% (6/16) of patients with paresthesia and cognition-related AEs, respectively, had a reduction in their DCP dose
 - Paresthesia AEs resolved in 5 of the 6 cases and cognition-related AEs resolved in all 6 cases
- Onset of paresthesia and cognition-related AEs typically occurred during the first 4 weeks of DCP treatment

Table 3. Characterization of Paresthesia and Cognition-Related Adverse Events During DCP Treatment

Parameter	Paresthesia AEs (N=63), n (%)	Cognition-Related AEs (N=63), n (%)
Any AE	25 (39.7)	16 (25.4)
Any drug-related AE*	24 (38.1)	16 (25.4)
Discontinuation due to AE	1 (1.6)	4 (6.3)
AE intensity		
Mild	11 (17.5)	9 (14.3)
Moderate	14 (22.2)	5 (7.9)
Severe	0 (0.0)	2 (3.2)
Resolution of last AE		
Resolved	19 (30.2)	11 (17.5)
Under treatment/observation	6 (9.5)	5 (7.9)

*Considered by investigators to be at least possibly related to study drug.
AE = adverse event; DCP = dichlorphenamide.

Conclusions

- **Long-term (61 weeks) DCP treatment was efficacious and provided durable reduction in attack frequency in patients with PPP**
- **Paresthesia and cognition-related AEs tended to be mild or moderate in intensity and nearly always first occurred early (eg, within 1 month) after therapy initiation**
- **These AEs rarely resulted in discontinuation from the study and were sometimes managed by DCP dose reductions**
- **Reduction in dose was frequently associated with resolution of these events, suggesting a potential intervention to hasten resolution**

References

1. Statland JM, et al. *Muscle Nerve*. 2018;57(4):522-530. 2. Cannon SC. *Compr Physiol*. 2015;5(2):761-790. 3. Sansone VA, et al. *Neurology*. 2016;86(15 Suppl):1408-1416. 4. Tawil R, et al. *Ann Neurol*. 2000;47(1):46-53. 5. Keveyis® (dichlorphenamide) tablets, for oral use [package insert]. Haifa Bay, Israel: Taro Pharmaceuticals Industries Ltd.; 2017.

Acknowledgements

The current analyses were supported by Strongbridge Biopharma, Trevose, PA, USA. The original study was supported, in part, by NIH National Institute of Neurological Disorders and Stroke grant R01 NS045686 and FDA Orphan Drug Program (FD-R-000821). Medical writing and technical editorial assistance was provided under direction of the authors by Mary Beth Moncrief, PhD, and Sophie Bolick, PhD, Synchrony Medical Communications, LLC, West Chester, PA. Funding for this support was provided by Strongbridge Biopharma.

Disclosures

NEJ reports receiving research support from Biogen Idec, Ionis Pharmaceuticals, Muscular Dystrophy Association, Myotonic Dystrophy Foundation, and Valerion Therapeutics; and serving as a consultant for AMO Pharma, Fulcrum Therapeutics, and Strongbridge Biopharma. MGH reports having nothing to disclose. JMS reports receiving research support from FSH Society, Muscular Dystrophy Association, and the National Institutes of Health; and serving as a consultant for Acceleron Pharma, Inc., Expansion Therapeutics, Fulcrum Therapeutics, Genea Biocells, and Strongbridge Biopharma. FC is an employee of Strongbridge Biopharma. RCG reports receiving research support from the Muscular Dystrophy Association, the National Institutes of Health, and Parent Project Muscular Dystrophy; and serving as a consultant for Idera, Medpace, PTC Therapeutics Inc., Sarepta Therapeutics Inc., Stealth BioTherapeutics, and Strongbridge Biopharma.

Annual Meeting of the American Association of Neuromuscular and Electrodiagnostic Medicine (AANEM) • Austin, TX • October 16–19, 2019

STRONGBRIDGE BIOPHARMA