Levoketoconazole in the Treatment of Endogenous Cushing's Syndrome: Secondary Endpoint Results From a Phase 3 Study (SONICS)

Maria Fleseriu, MD[1]; Rosario Pivonello, MD[2]; Atanaska Elenkova, MD, PhD[3]; Roberto Salvatori, MD[4]; Richard Auchus, MD, PhD[5]; Richard A. Feelders, MD, PhD[6]; Eliza B. Geer, MD[7]; Yona Greenman, MD[8]; Przemyslaw Witek, MD, PhD[9]; Fredric Cohen, MD[10]; Beverly M.K. Biller, MD[11]

[1]Oregon Health and Science University, Portland, OR, USA; [2]University of Naples Federico II, Naples, Italy; [3]Medical University Sofia, Sofia, Bulgaria; [4]Johns Hopkins University, Baltimore, MD, USA; [5]University of Michigan Medical School, Ann Arbor, MI, USA; [6]Erasmus Medical Center, Rotterdam, The Netherlands; [7]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [8]Tel Aviv University, Tel Aviv, Israel; [9]Military Institute of Medicine, Warsaw, Poland; [10]Strongbridge Biopharma, Trevose, PA, USA; [11]Massachusetts General Hospital, Boston, MA, USA

Poster #12

Introduction

- Levoketoconazole, a ketoconazole stereoisomer, is a steroidogenesis inhibitor in development for the treatment of endogenous Cushing's syndrome (CS), a rare endocrine disease characterized by overproduction of cortisol[1,2]

Objective

- To assess the safety and efficacy of levoketoconazole in the treatment of endogenous CS, with a focus on selected secondary endpoints related to cortisol measurements

Methods

Study Design and Patients

- The Study of levOketocoNazole In Cushing's Syndrome (SONICS) is a phase 3, single-arm, open-label, dose-titration study of oral levoketoconazole in adults with confirmed diagnosis of CS and elevated mean 24-hour urinary free cortisol (mUFC) levels ≥1.5× upper limit of normal (ULN; calculated from ≥4 adequately collected samples; **Figure 1**)

Figure 1. Study Design

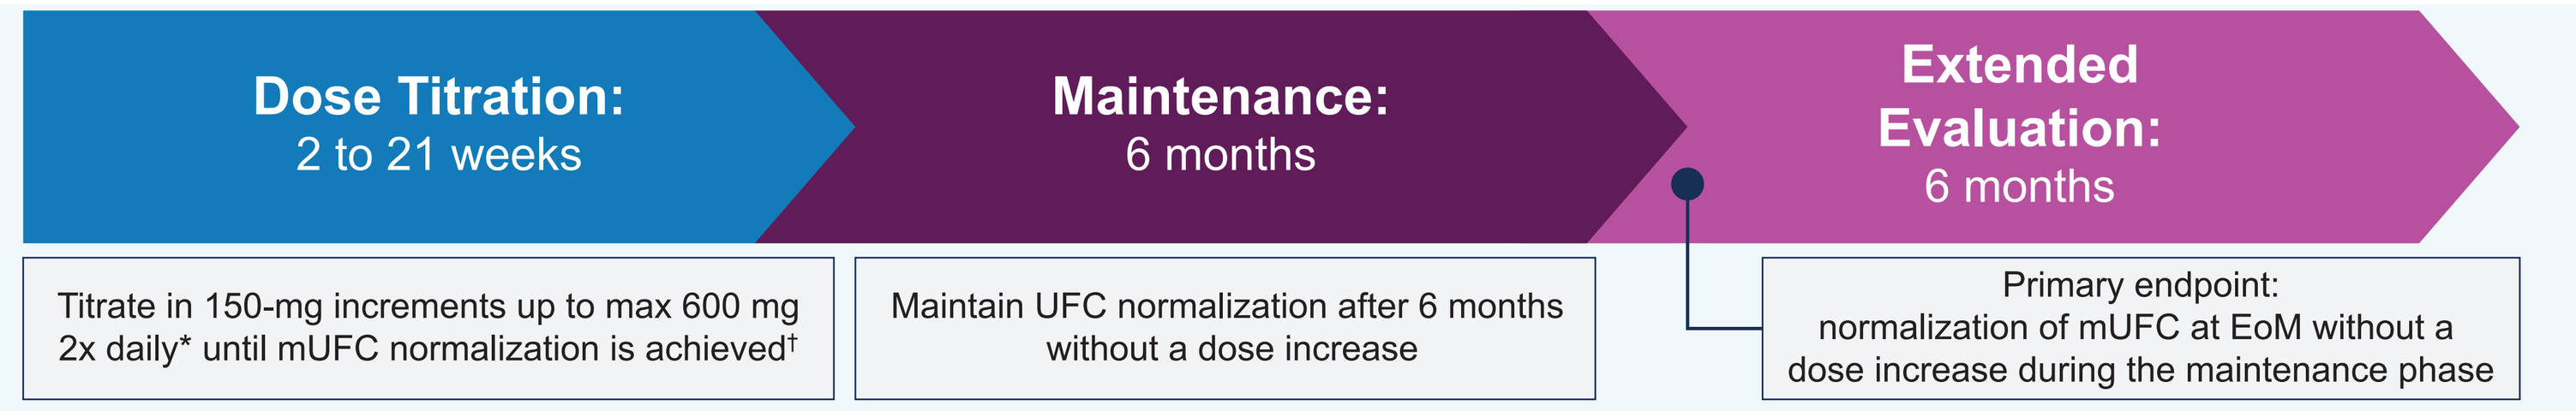
Dose Titration:
2 to 21 weeks
Maintenance:
6 months
Extended Evaluation:
6 months
Titrate in 150-mg increments up to max 600 mg 2x daily* until mUFC normalization is achieved†
Maintain UFC normalization after 6 months without a dose increase
Primary endpoint: normalization of mUFC at EoM without a dose increase during the maintenance phase

*All patients started at the protocol-mandated 150 mg twice daily, but a reduction to 150 mg once daily to improve tolerability was allowed. †A therapeutic dose was considered established when either: [a] mean UFC levels were ≤ULN or [b] a maximum allowed dose of 600 mg twice daily or [c] a maximal tolerated dose had been reached and there was a clinically meaningful partial response in the opinion of the investigator.
EoM = end of maintenance; mUFC = mean urinary free cortisol; ULN = upper limit of normal.

Outcomes

- Primary endpoint: mUFC normalization (mUFC ≤ULN) at end of maintenance (EoM) was presented previously[3]
- Select secondary endpoints reported here (through EoM):
 - Percentage of complete responders at all monthly visits in maintenance phase in the overall intent-to-treat (ITT) population
 - Changes from baseline in mUFC and late night salivary cortisol (LNSC) levels in the overall maintenance population and in a subset of the maintenance population with Cushing's disease (CD)
 - Changes from baseline in adrenocorticotrophic hormone (ACTH) levels in the subset of the maintenance population with CD
 - Pituitary tumor size was measured at EoM, but results are not available for this presentation
- Safety: incidence of treatment-emergent adverse events (AEs); prespecified AEs of special interest (potential liver toxicity, QTc prolongation, and adrenal insufficiency) through EoM

Results

Patient Population

- Ninety-four patients were enrolled and received ≥1 dose of study medication (ITT population; **Table 1**, **Figure 2**)

Figure 2. Patient Disposition

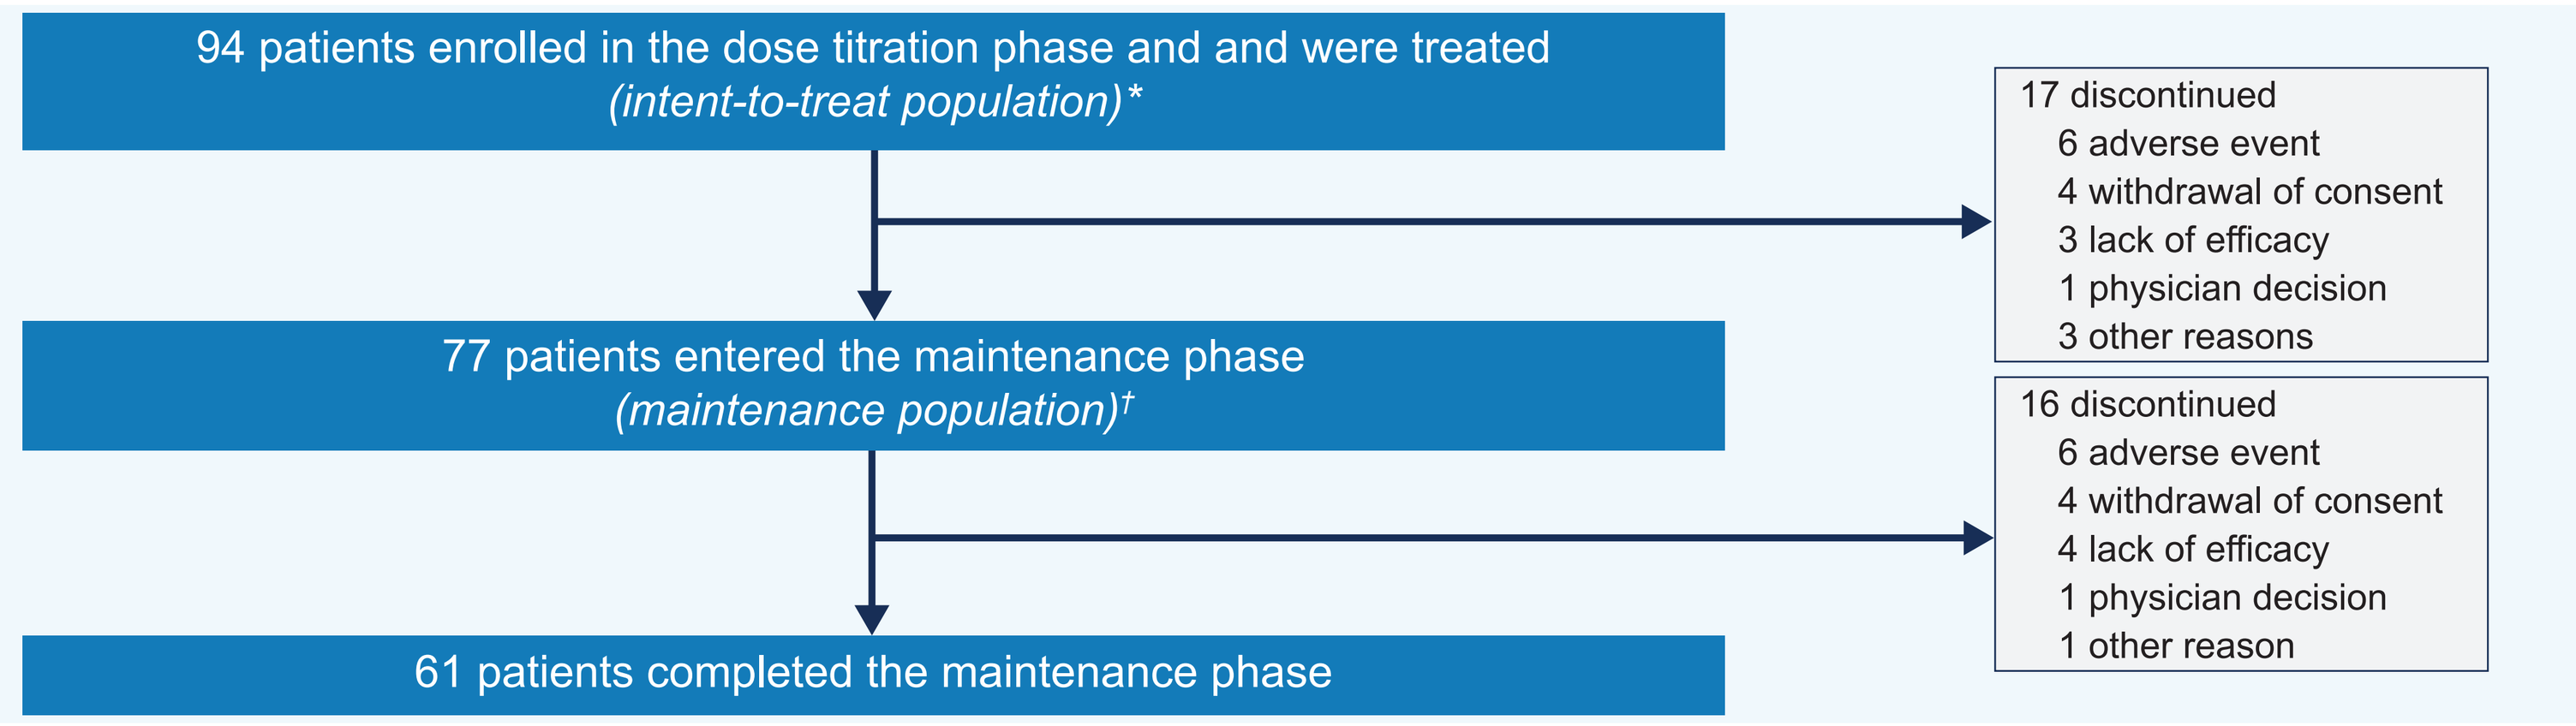
94 patients enrolled in the dose titration phase and and were treated
(intent-to-treat population)*
17 discontinued
6 adverse event
4 withdrawal of consent
3 lack of efficacy
1 physician decision
3 other reasons
77 patients entered the maintenance phase
(maintenance population)†
16 discontinued
6 adverse event
4 withdrawal of consent
4 lack of efficacy
1 physician decision
1 other reason
61 patients completed the maintenance phase

*Intent-to-treat population included all patients who received ≥1 dose of study medication. †Maintenance population consisted of all patients who entered the maintenance phase and received ≥1 dose of study medication during this phase.

Table 1. Demographic and Baseline Characteristics (ITT Population)

Characteristic	Patients (N=94)
Age, y, mean (SD)	43.7 (13.4)
Female, n (%)	77 (81.9)
Race, n (%)	
White	90 (95.7)
Black	1 (1.1)
Other	1 (1.1)
Unknown	2 (2.1)
Weight, kg, mean (SD)	84.0 (23.4)
BMI, kg/m^2,* mean (SD)	30.8 (8.2)
Time since Cushing's syndrome diagnosis, mo, mean (SD)	68.0 (80.4)
Diagnosis of Cushing's disease, n (%)	80 (85.1)
Diabetes, n (%)	36 (38.3)
Hypertension, n (%)	67 (71.3)
Hypercholesterolemia, n (%)	34 (36.2)
Baseline mUFC,†‡ nmol/24 h; μg/24 h	
Mean (SD)	671.4 (743.1); 243.3 (269.2)
Median (range)	407.9 (162.0–4168.0); 147.8 (58.7–1510.1)
Baseline mUFC,‡ × ULN§	
Mean (SD)	4.9 (5.4)
Median (range)	3.0 (1.2–30.2)¶

*n=93; 1 patient had missing BMI data due to missing height information. †For each patient, the average of the UFCs from the adequate samples at baseline were calculated. ‡n=92; 2 patients did not have ≥2 adequate urine samples at baseline. §ULN for UFC = 138 nmol/24 hours (50 μg/24 hours). ¶1 patient with mUFC <1.5× ULN owing to inadequate urine collection.
BMI = body mass index; ITT = intent-to-treat; mUFC = mean urinary free cortisol; SD = standard deviation; ULN = upper limit of normal.

Efficacy

- Percentage of mUFC complete responders (ie, mUFC normalizers without a dose increase during the maintenance phase) ranged from 30% to 50% during monthly visits in the maintenance phase (**Figure 3**)

Figure 3. Percentage of Complete Responders From Baseline Through Month 6 (ITT Population)*†

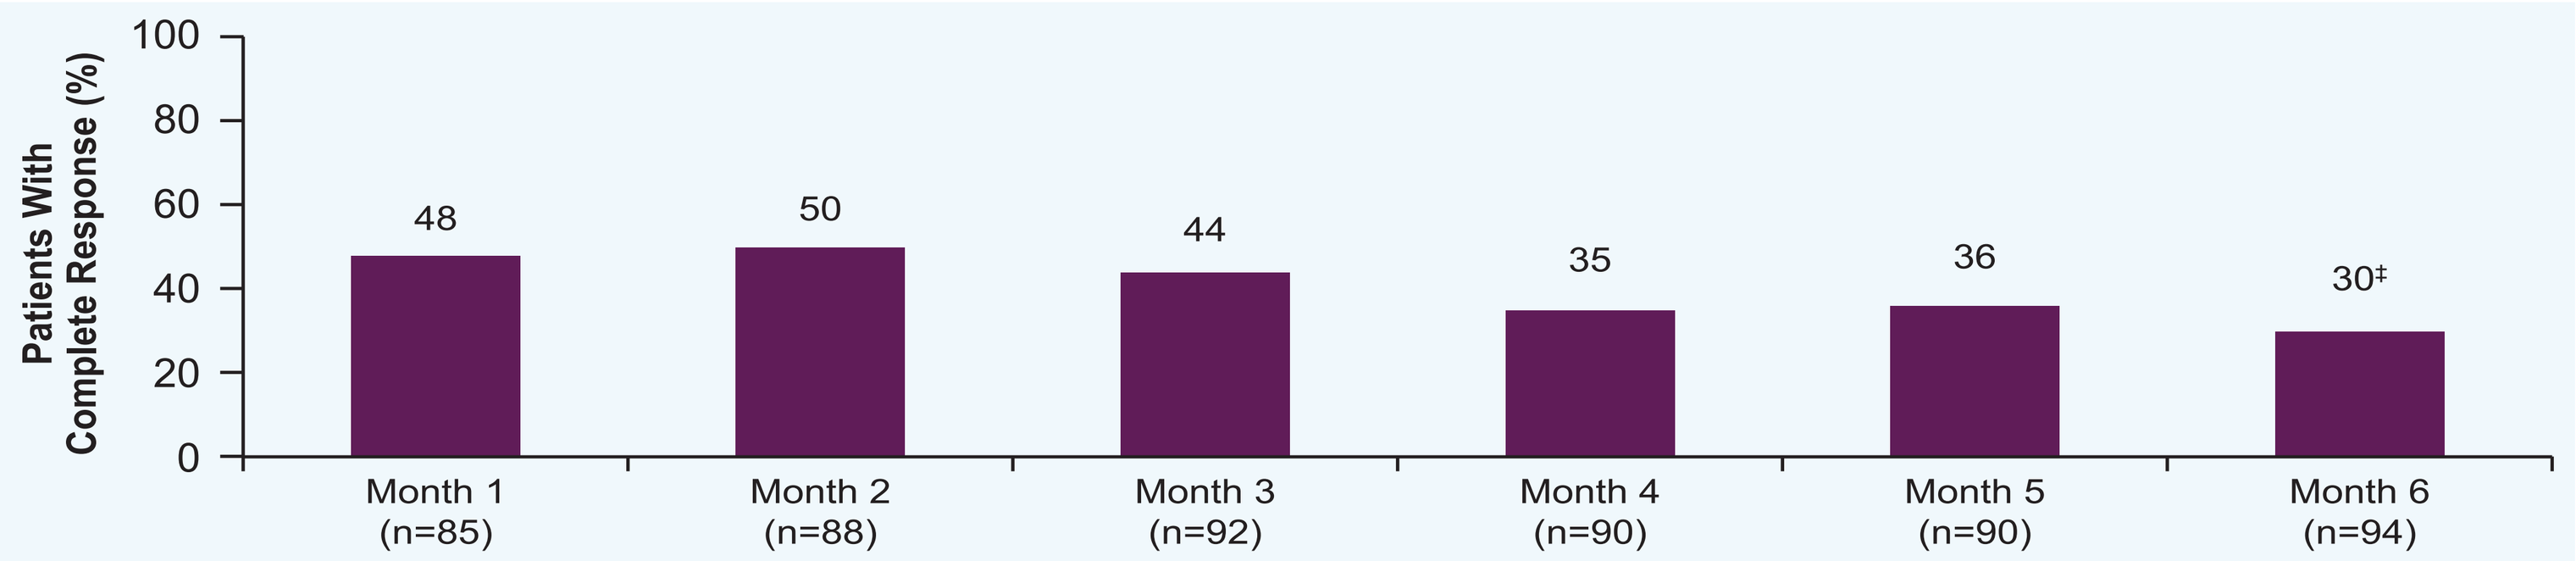
Patients With Complete Response (%)
100
80
60
40
20
0
48
50
44
35
36
30‡
Month 1 (n=85)
Month 2 (n=88)
Month 3 (n=92)
Month 4 (n=90)
Month 5 (n=90)
Month 6 (n=94)

*Complete response defined as mUFC ≤ULN (ULN for UFC = 138 nmol/24 hours [50 μg/24 hours]) without a dose increase. †There was no data imputation, except as follows: a patient with dose increase during the maintenance phase was imputed as a non-responder from the first visit following the dose increase to Month 6. Missing data at Month 6 was imputed as a non-response. ‡Response rate was statistically significant as compared to the null hypothesis of 20% or lower rate at Month 6. One-sided *P* value of 0.0154 was obtained from a mixed-effects repeated measures model with an underlying binomial distribution and adjustments for covariates.
ITT = intent-to-treat; mUFC = mean urinary free cortisol; ULN = upper limit of normal.

- Mean mUFC levels decreased significantly from baseline to Month 1 visit (Day 30 of maintenance phase) and remained lower than baseline through EoM (**Figure 4**)

Figure 4. Mean Change From Baseline in mUFC Levels Through Month 6 (Maintenance Population)

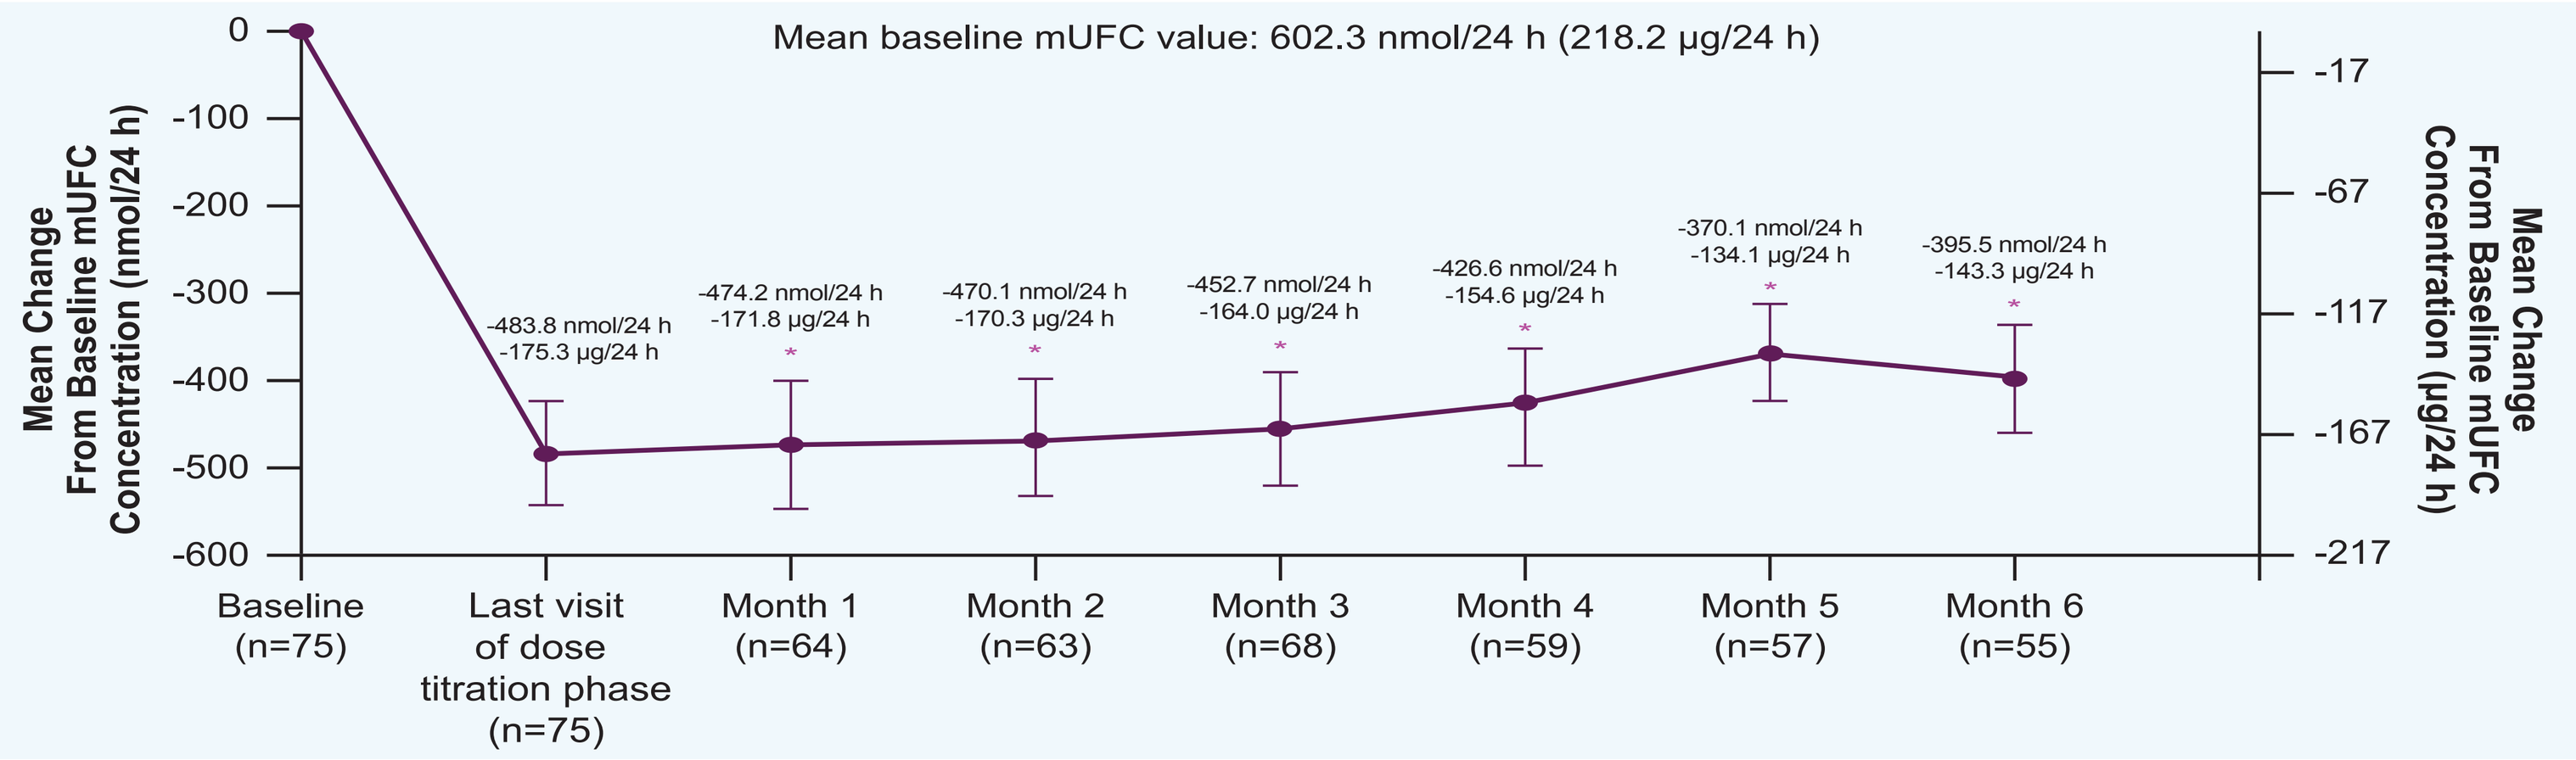
Mean baseline mUFC value: 602.3 nmol/24 h (218.2 μg/24 h)
Mean Change From Baseline mUFC Concentration (nmol/24 h)
Mean Change From Baseline mUFC Concentration (μg/24 h)
0
-100
-200
-300
-400
-500
-600
-17
-67
-117
-167
-217
-483.8 nmol/24 h
-175.3 μg/24 h
-474.2 nmol/24 h
-171.8 μg/24 h
-470.1 nmol/24 h
-170.3 μg/24 h
-452.7 nmol/24 h
-164.0 μg/24 h
-426.6 nmol/24 h
-154.6 μg/24 h
-370.1 nmol/24 h
-134.1 μg/24 h
-395.5 nmol/24 h
-143.3 μg/24 h
Baseline (n=75)
Last visit of dose titration phase (n=75)
Month 1 (n=64)
Month 2 (n=63)
Month 3 (n=68)
Month 4 (n=59)
Month 5 (n=57)
Month 6 (n=55)

Two-sided *P* value from the paired t-test performed on the change from baseline to each maintenance phase visit. Error bars represent standard error of the mean. *$P<0.0001$.
mUFC = mean urinary free cortisol.

- More patients in the non-responder group had an initial response to treatment that left mUFC >ULN compared with the complete responder group, whereas the groups had relatively similar maintenance of their initial mUFC response over time (**Figure 5**)

Figure 5. Individual Line Plots of mUFC (× ULN) by Response

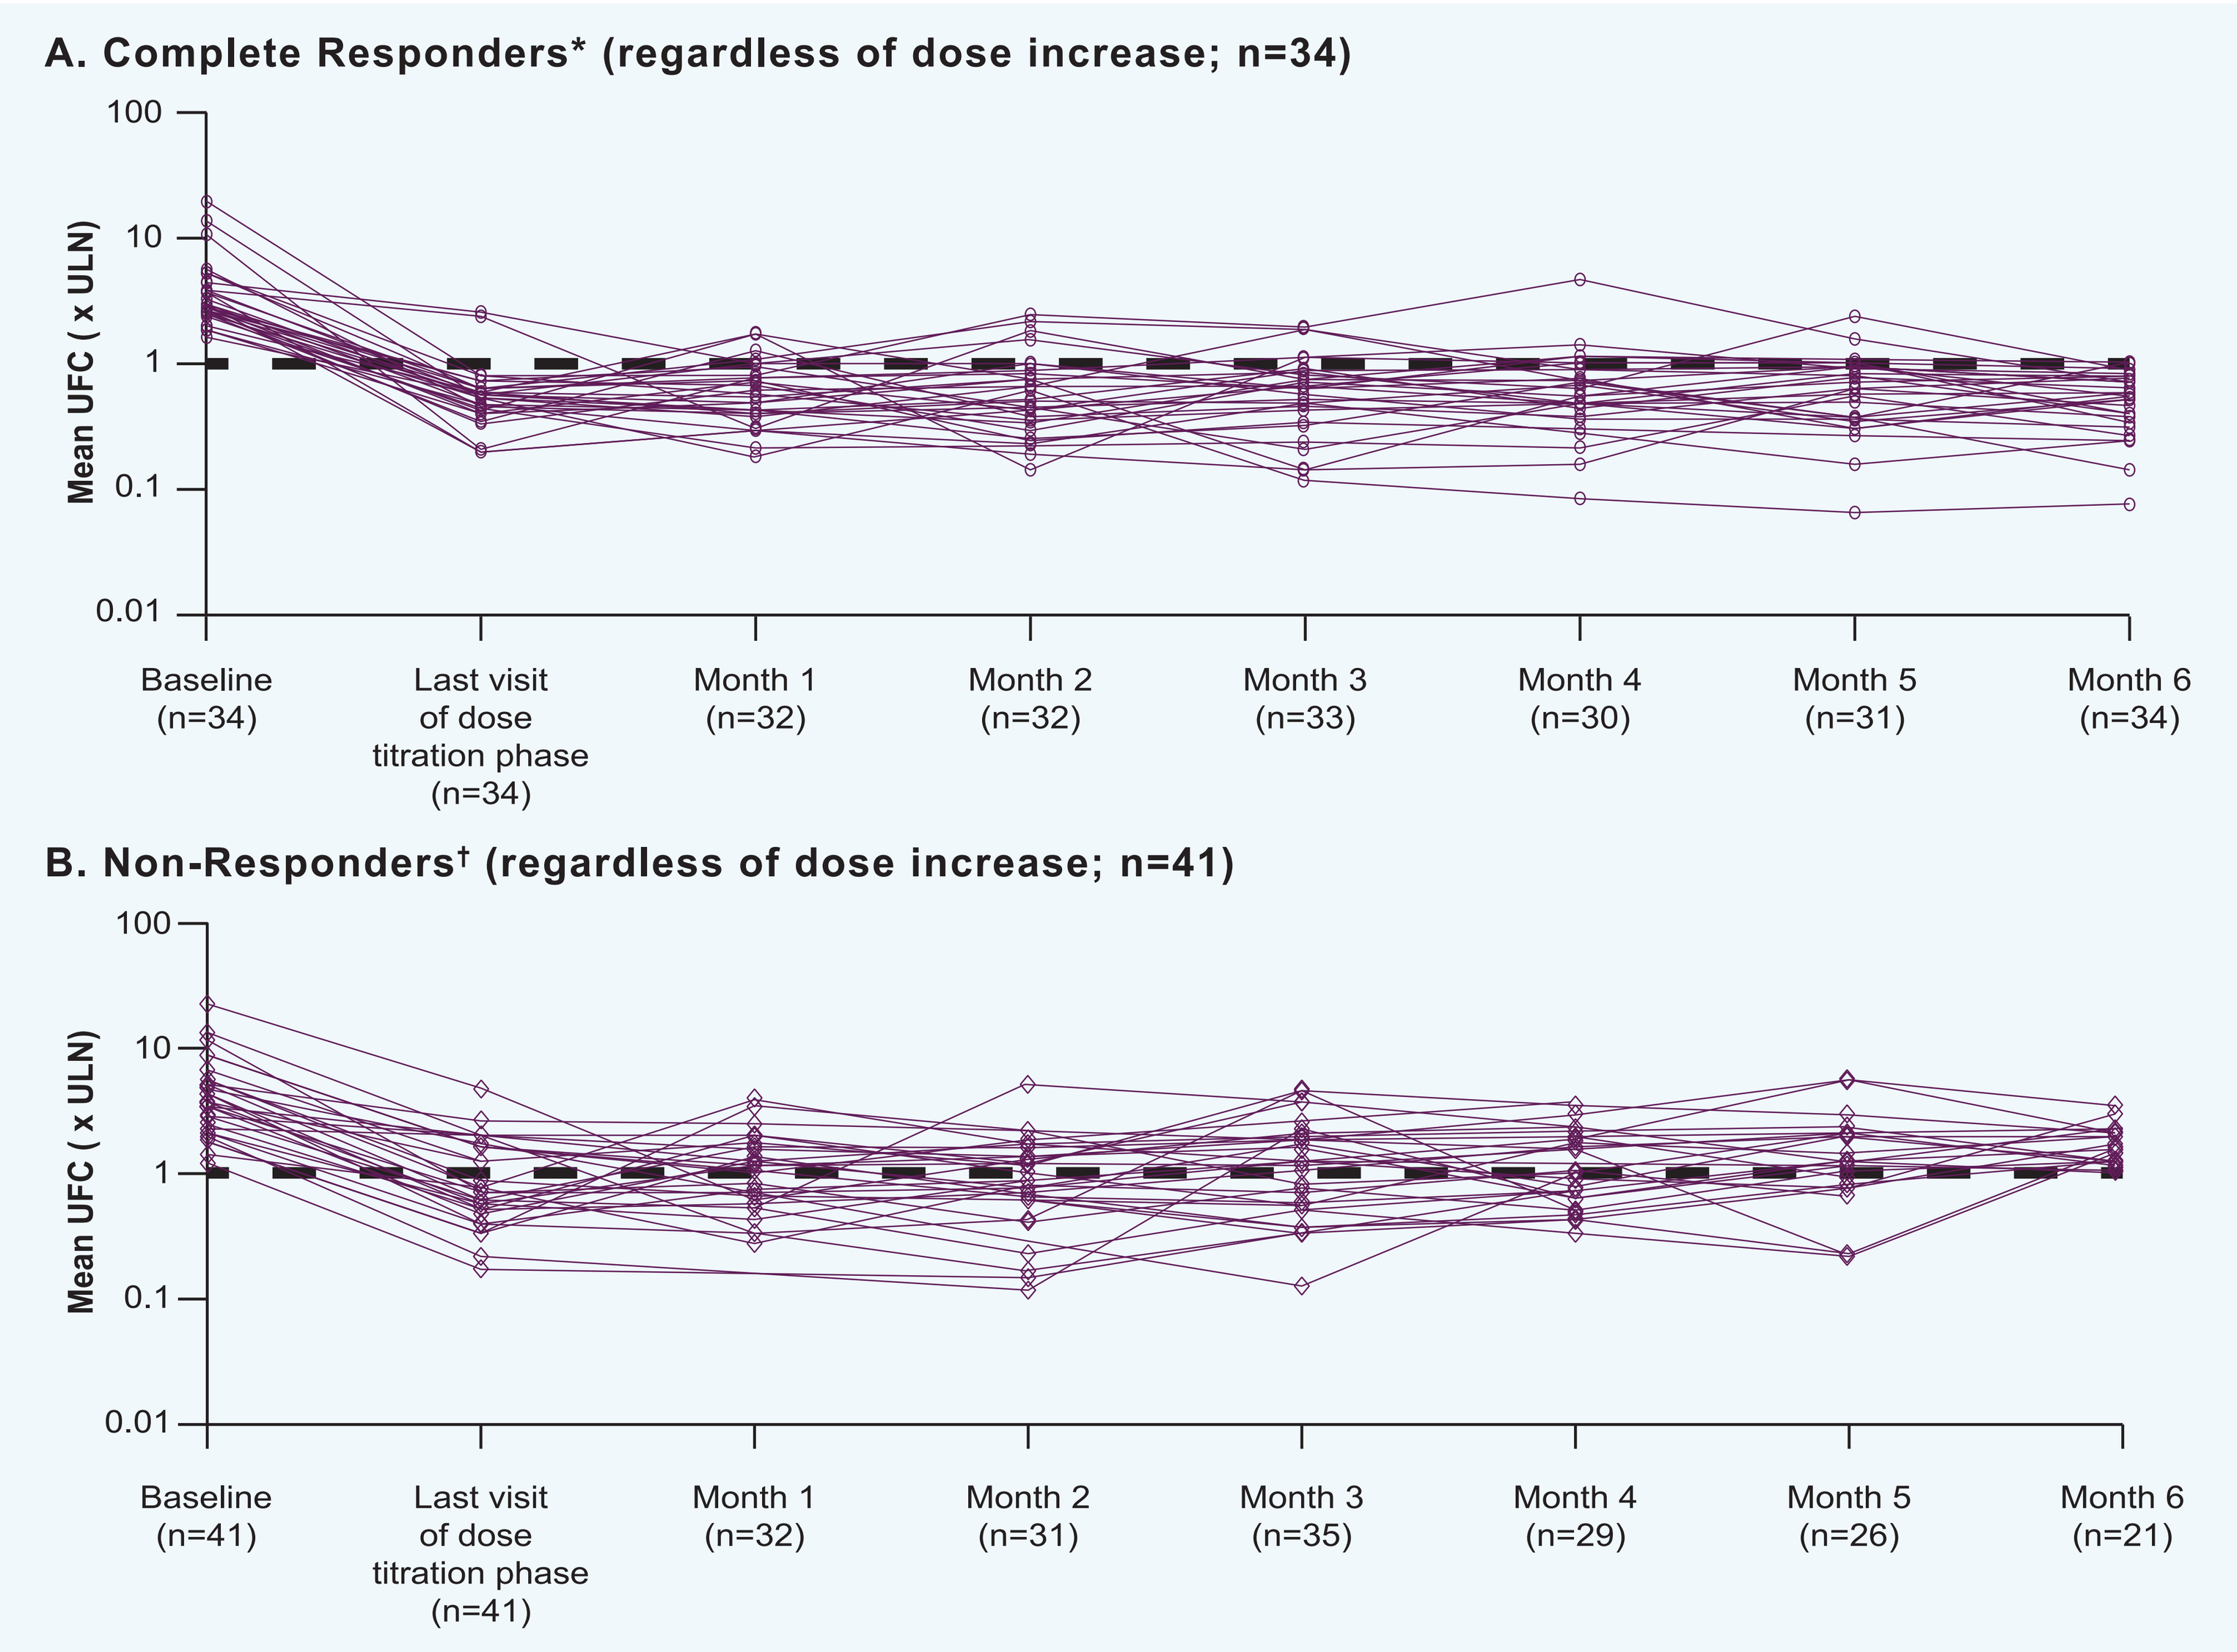
A. Complete Responders* (regardless of dose increase; n=34)
Mean UFC (x ULN)
100
10
1
0.1
0.01
Baseline (n=34)
Last visit of dose titration phase (n=34)
Month 1 (n=32)
Month 2 (n=32)
Month 3 (n=33)
Month 4 (n=30)
Month 5 (n=31)
Month 6 (n=34)
B. Non-Responders† (regardless of dose increase; n=41)
Mean UFC (x ULN)
100
10
1
0.1
0.01
Baseline (n=41)
Last visit of dose titration phase (n=41)
Month 1 (n=32)
Month 2 (n=31)
Month 3 (n=35)
Month 4 (n=29)
Month 5 (n=26)
Month 6 (n=21)

Two patients had no non-missing mUFC in the maintenance phase. They were considered as non-responders but are not represented in this figure. The dashed line represents the ULN for urinary free cortisol (138 nmol/24 hours [50 μg/24 hours]). *Complete response defined as mUFC ≤ULN at EoM without a dose increase. †Non-response: mUFC >ULN at EoM.
EoM = end of maintenance; mUFC = mean urinary free cortisol; ULN = upper limit of normal.

- Similar to mUFC, LNSC levels decreased significantly from baseline to Month 1 visit of maintenance phase and remained lower than baseline through EoM (**Figure 6**)
 - In both the overall maintenance population and in the subset of patients with CD, LNSC levels uncommonly normalized (percentage of patients with high LNSC at baseline that became normal [≤2.5 nmol/L (0.09 μg/dL)] over time ranged from 4% to 18% in the overall population and 5% to 11% in the subset of patients with CD)

Figure 6. Change From Baseline in LNSC Levels Through Month 6 (Maintenance Population)

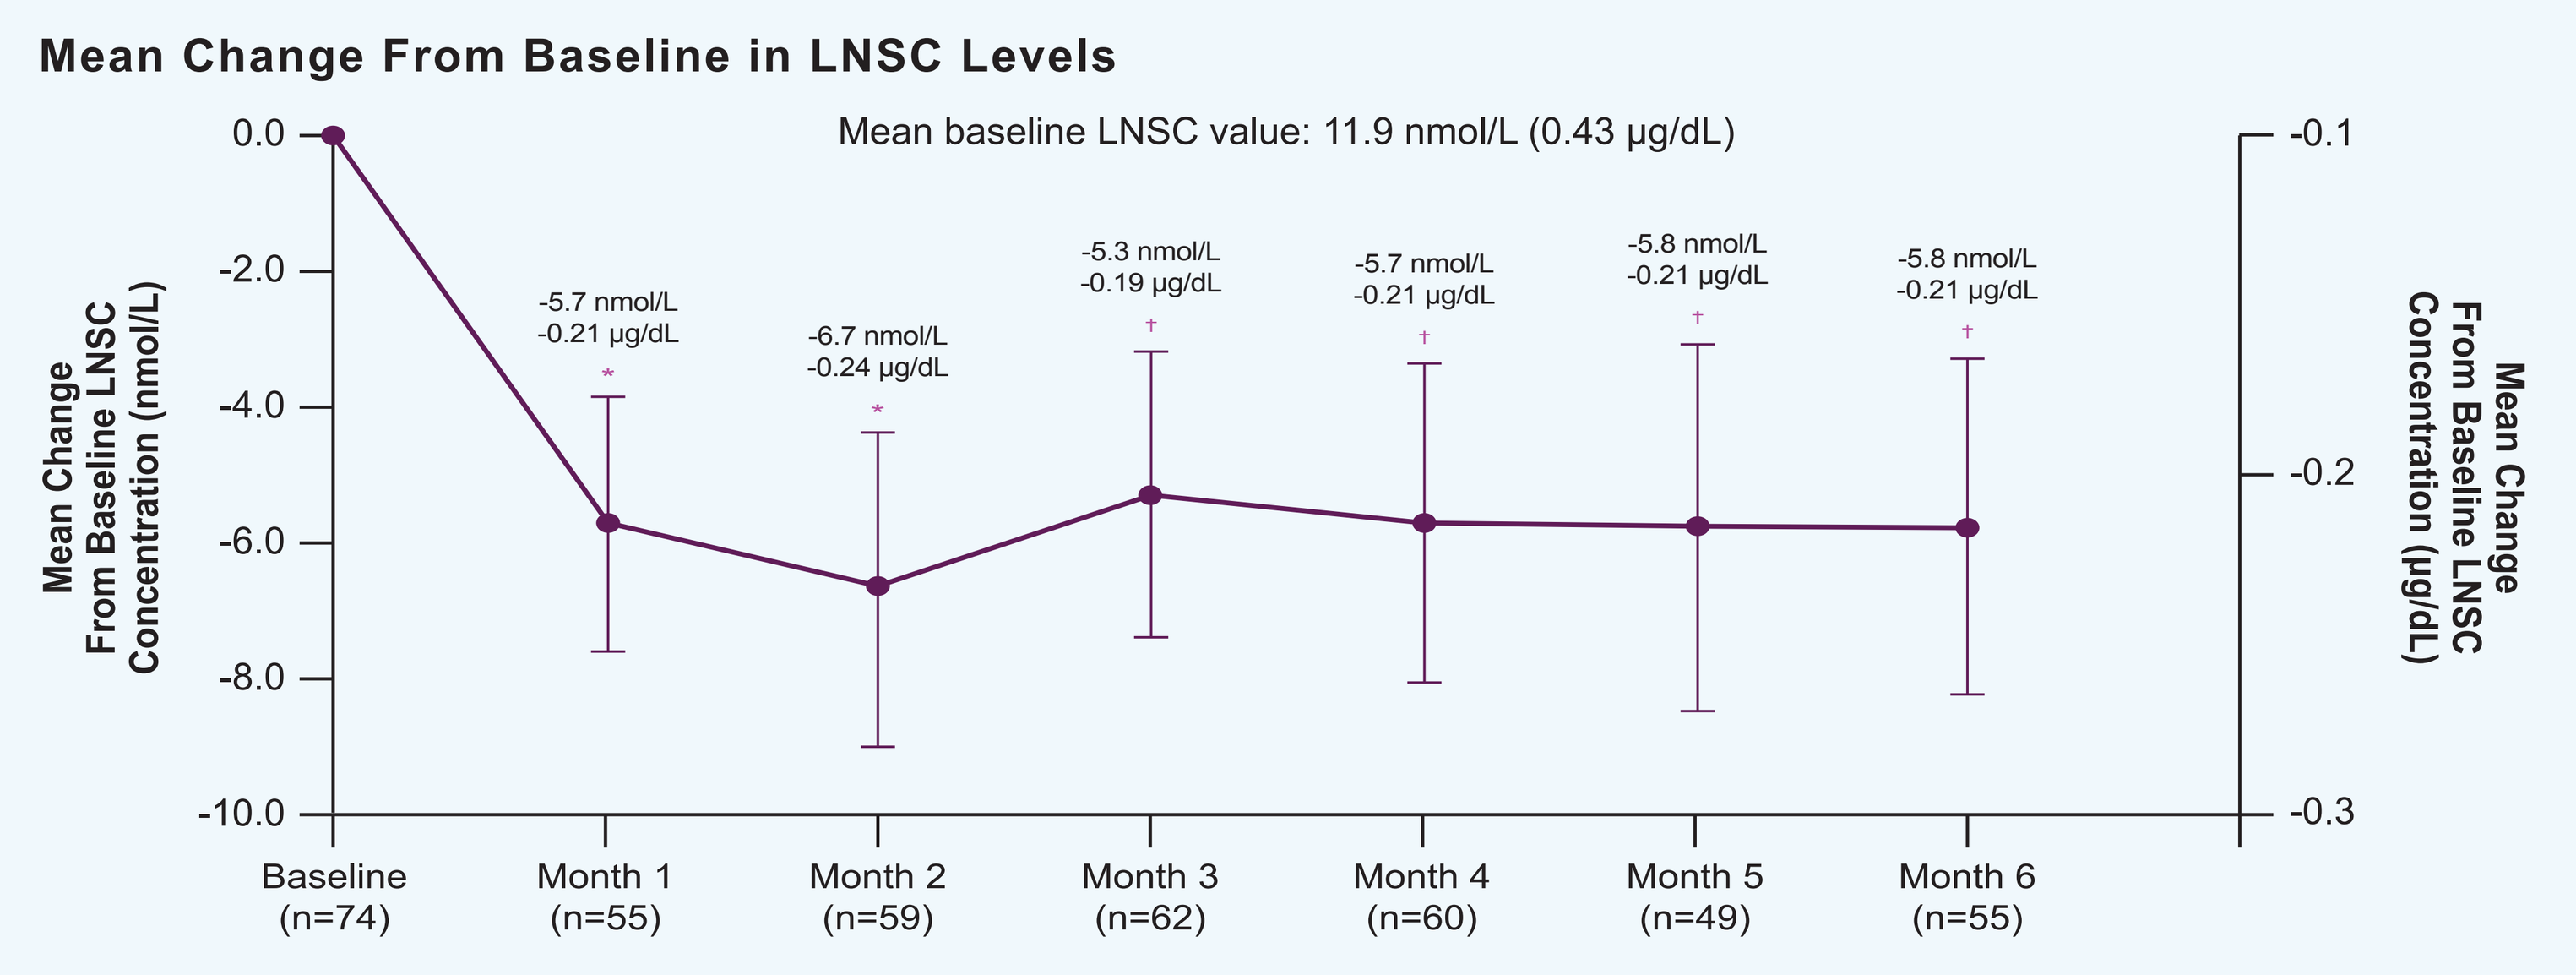
Mean Change From Baseline in LNSC Levels
Mean baseline LNSC value: 11.9 nmol/L (0.43 μg/dL)
Mean Change From Baseline LNSC Concentration (nmol/L)
Mean Change From Baseline LNSC Concentration (μg/dL)
0.0
-2.0
-4.0
-6.0
-8.0
-10.0
-0.1
-0.2
-0.3
-5.7 nmol/L
-0.21 μg/dL
-6.7 nmol/L
-0.24 μg/dL
-5.3 nmol/L
-0.19 μg/dL
-5.7 nmol/L
-0.21 μg/dL
-5.8 nmol/L
-0.21 μg/dL
-5.8 nmol/L
-0.21 μg/dL
Baseline (n=74)
Month 1 (n=55)
Month 2 (n=59)
Month 3 (n=62)
Month 4 (n=60)
Month 5 (n=49)
Month 6 (n=55)

Two-sided *P* value from the paired t-test performed on the change from baseline to each maintenance phase visit. Error bars represent standard error of the mean. *$P<0.01$. †$P<0.05$.
LNSC = late night salivary cortisol.

- In the subset of patients with CD, mean ACTH level at baseline was 1.5× ULN and at end of maintenance was 2.9× ULN; 61% of 46 patients with available data had ACTH levels more than 2× ULN at the end of maintenance

Safety

- The most commonly reported AEs were nausea, headache, and peripheral edema (**Table 2**)

Table 2. Summary of Adverse Events (Both Phases; ITT Population)

Adverse Event, n (%)	Patients (N=94)
Any AE	92 (97.9)
Serious AE	14 (14.9)
Drug-related AE*	40 (42.6)
AE leading to discontinuation	12 (12.8)
Intensity of AEs	
Mild	21 (22.3)
Moderate	54 (57.4)
Severe	15 (16.0)
Life threatening	1 (1.1)
Death†	1 (1.1)
Most common AEs‡	
Nausea	30 (31.9)
Headache	26 (27.7)
Peripheral edema	18 (19.1)
Hypertension	16 (17.0)
Fatigue	15 (16.0)
Diarrhea	14 (14.9)
ALT increased	14 (14.9)

*AEs that were assessed by the investigator as probably or definitely related to study drug. †Patient died of colon cancer; unrelated to study drug. ‡Reported in ≥14% of patients.
AE = adverse event; ALT = alanine aminotransferase; ITT = intent-to-treat.

- Serious AEs were reported in 14 patients, of which 4 were considered to be probably or definitely related to levoketoconazole: elevated liver function test results (1 patient), prolonged QTc interval (2 patients), and adrenal insufficiency (1 patient)
- In all 10 patients (11%) with alanine aminotransferase (ALT) >3× ULN (including 3% with ALT >5× ULN), the ALT elevation was reversible without clinical sequelae
- Nine patients (10%) had ≥1 QTc value representing an increase of >60 msec from baseline, and 2 patients (2%) had ≥1 confirmed QTc value of >500 msec; all QTc prolongations were reversed with temporary drug interruption, and all patients affected were able to resume levoketoconazole at the same or lower dose following the prolongation event

Conclusions

- **In this large, international, prospective trial of endogenous CS, levoketoconazole treatment led to sustained reductions in both mUFC and LNSC levels over a 6-month maintenance treatment period**
- **LNSC was less commonly normalized than was mUFC, indicating improvement in but generally incomplete normalization of cortisol diurnal rhythmicity**
- **The lower proportion of patients normalizing mUFC after Month 2 of maintenance phase largely reflects incomplete normalization early in therapy in a subset of treated patients rather than an escape phenomenon in a subset of patients (Figures 5A and 5B)**
- **ACTH levels in patients with CD increased less than 2-fold from baseline to EoM**
- **Levoketoconazole was generally well tolerated with 13% discontinuing at any time due to an AE; there were no unexpected safety signals observed through EoM**

References

1. Creemers SG, et al. *Expert Opin Pharmacother*. 2015;16(12):1829-1844. 2. Fleseriu M, Castinetti F. *Pituitary*. 2016;19:643-653. 3. Fleseriu M, et al. Presented at the 18th Congress of the European NeuroEndoCrine Association; October 17-20, 2018; Wroclaw, Poland.

Acknowledgements

This study was funded by Cortendo AB (a subsidiary of Strongbridge Biopharma, Trevose, PA, USA). The authors acknowledge all Investigators, Sub-Investigators, and Research Nurses/Study Coordinators who performed the study and all patients who participated in the study. Medical editorial assistance was provided by Pratibha Hebbar, PhD, of Synchrony Medical Communications, LLC, West Chester, PA. Funding for this support was provided by Strongbridge Biopharma.

Disclosures

MF reports receiving research grants to OHSU from Millendo Therapeutics, Novartis, and Strongbridge Biopharma; and serving as a consultant for Novartis and Strongbridge Biopharma. RP reports serving as the principal investigator of research grants to Federico II University from Corcept Therapeutics, Novartis, and Strongbridge Biopharma; and receiving consulting honoraria from Novartis and Strongbridge Biopharma. AE reports receiving consulting honoraria from Novartis. RS reports serving as the principal investigator of research grants to Johns Hopkins University from Novartis and Strongbridge Biopharma. RA reports receiving research grants to the University of Michigan from Novartis and Strongbridge Biopharma; and serving as a consultant for Novartis and Strongbridge Biopharma. RF reports receiving research grants from Novartis; and serving on the speakers' bureau for HRA Pharma and Novartis. EG reports receiving research grants to MSKCC for participation in clinical trials from Ionis, Novartis, and Strongbridge Biopharma; and serving as a scientific consultant for Corcept Therapeutics, Novartis, and Strongbridge Biopharma. YG reports serving as the principal investigator of research grants to Tel Aviv-Sourasky Medical Center from Chiasma, Novartis, and Strongbridge Biopharma; and receiving lecture fees from Medison, Novartis, and Pfizer. PW reports receiving travel grants from Ipsen and Novartis; and receiving personal fees as a clinical investigator from Ipsen, Novartis, Novo Nordisk, and Strongbridge Biopharma. FC reports being an employee of Strongbridge Biopharma. BB reports serving as the principal investigator of research grants to Massachusetts General Hospital from Novartis and Strongbridge Biopharma; and receiving consulting honoraria from Novartis and Strongbridge Biopharma.

STRONGBRIDGE BIOPHARMA

16th International Pituitary Congress; New Orleans, LA; March 20-22, 2019